Poster #C40

Comparison of Healthcare Resource Utilization and Total Direct Costs of Care Among Patients with Chronic Lymphocytic Leukemia/ Small Lymphocytic Lymphoma Following First-Line Treatment with Ibrutinib or Chemoimmunotherapy

Raisa Volodarsky, PharmD, RPh[1], Mukul Singhal, PhD[2], Joseph L. Smith, PhD, MPH[2], Reethi Iyengar, PhD, MBA, MHM[1], Swetha Challagulla, MS[1]

[1]Pharmacyclics LLC, an AbbVie Company, Sunnyvale, CA, USA; [2]HealthCore Inc., Wilmington, DE, USA

INTRODUCTION

- The Surveillance, Epidemiology, and End Results (SEER) Program estimated nearly 21,040 new cases and about 4,060 deaths due to chronic lymphocytic leukemia/small lymphocytic lymphoma (CLL/SLL) in 2020.[1]
- Ibrutinib is an oral, once-daily Bruton's tyrosine kinase (BTK) inhibitor that received FDA approval in March 2016 as a first-line (1L) treatment for CLL/SLL.
- Ibrutinib is the only targeted therapy to demonstrate both a significant progression-free survival[2-7] and overall survival[2,3,6,7] benefit versus chemotherapy and/or immunotherapy in multiple randomized phase 3 studies in both previously untreated and relapsed/refractory CLL/SLL.
- There is limited evidence in terms of healthcare resource utilization (HRU) and expenditure associated with patients with CLL/SLL using ibrutinib and chemoimmunotherapy (CIT) in the real-world setting.
- Several real-world studies have demonstrated HRU and cost benefit of 1L ibrutinib use in comparison with CIT in patients with CLL/SLL. The purpose of this study is to evaluate a previously unstudied commercially insured population and assess the generalizability of published economic outcomes among CLL/SLL patients treated with 1L ibrutinib and CIT.

OBJECTIVE

- To compare HRU and total direct cost of care in patients with CLL following initiation of 1L single-agent ibrutinib vs CIT using a large national commercial claims database

METHODS

Figure 1: Study Design

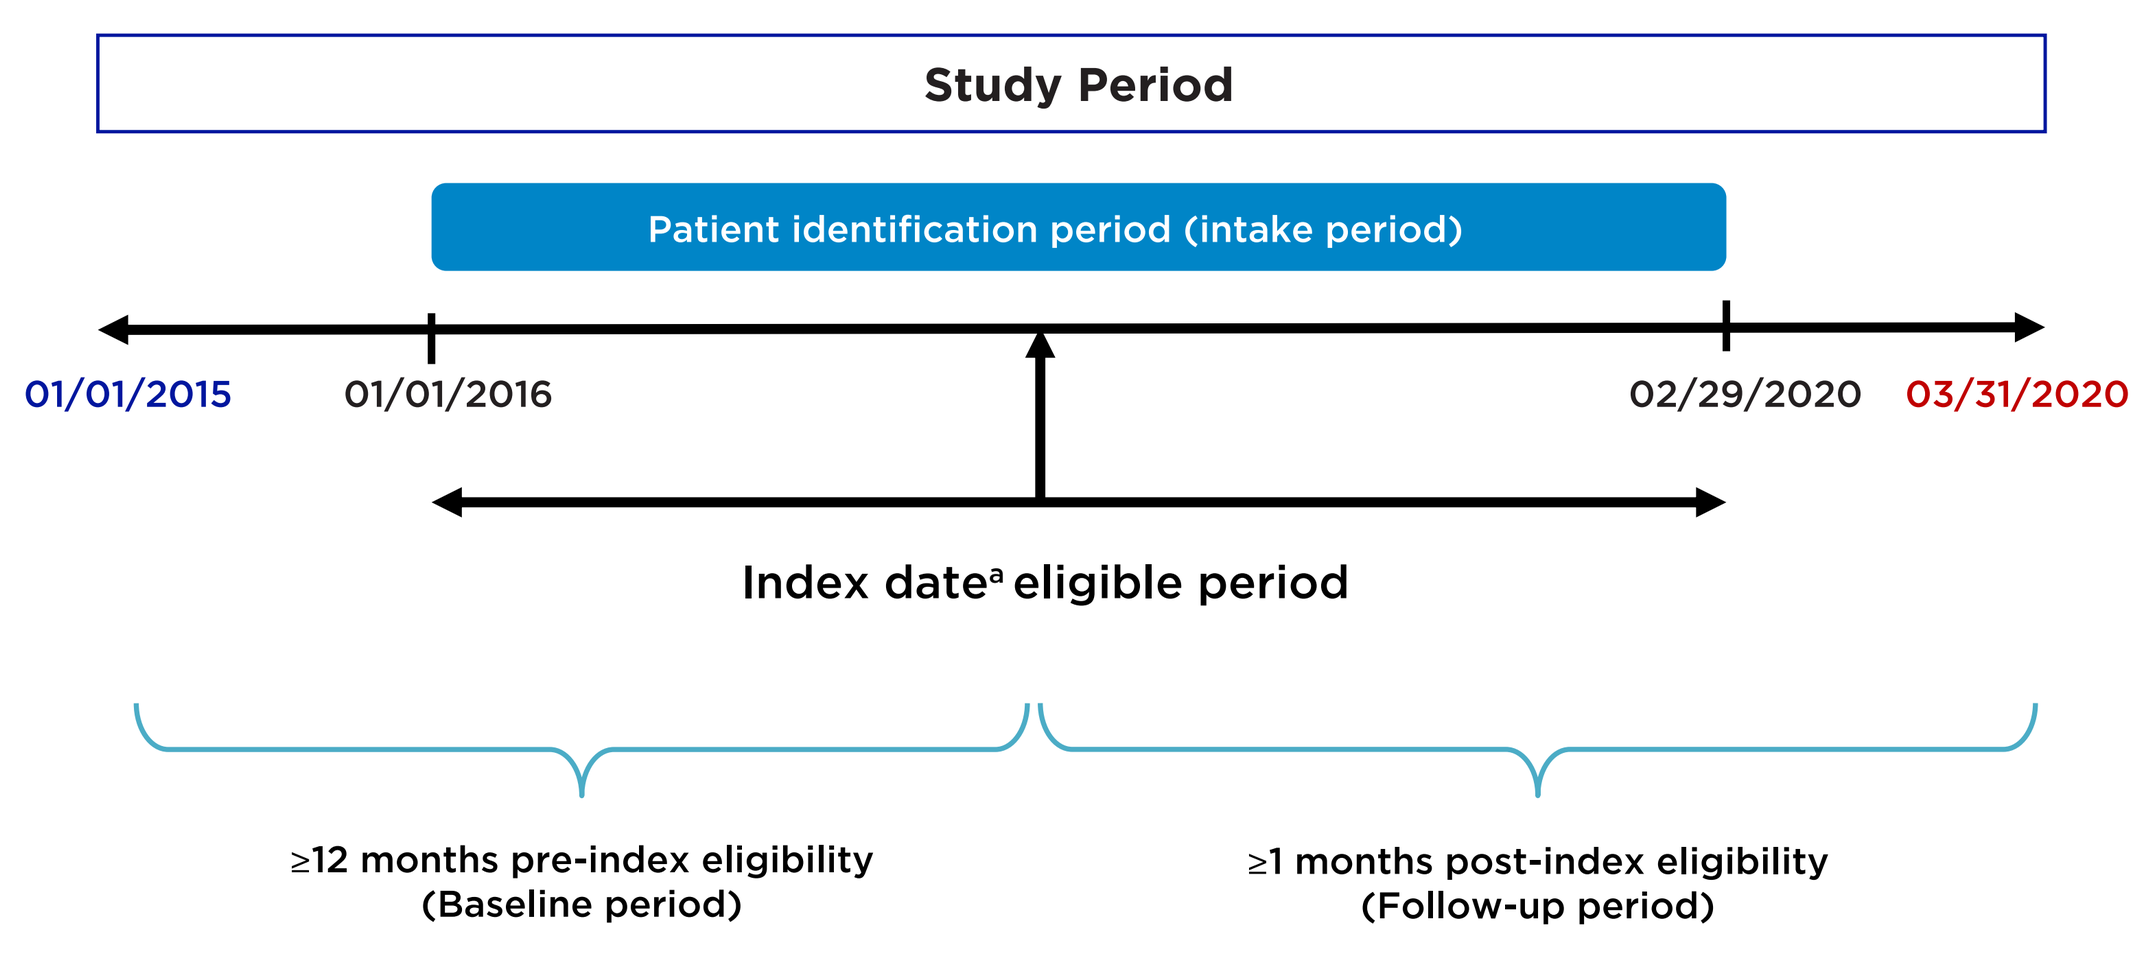
Study Period
Patient identification period (intake period)
01/01/2015
01/01/2016
02/29/2020
03/31/2020
Index date[a] eligible period
≥12 months pre-index eligibility (Baseline period)
≥1 months post-index eligibility (Follow-up period)

[a]All patients with at least one medical or pharmacy claim for ibrutinib or CIT between 01/01/2016 and 2/29/2020 (intake period) were identified. The earliest date with the claim for ibrutinib or CIT was set as the index date.

Study Design and Data Source (Figure 1)

- This study used a retrospective, observational medical and pharmacy claims-based, cohort study design, using the HealthCore Integrated Research Database (HIRD®) during the study period of 1/1/2015 to 3/31/2020.
- The HIRD contains integrated, longitudinal claims data on over 75 million people across 14 geographically diverse health plans across the US going back to 2006.
- Follow-up period was calculated as the duration of index treatment (ibrutinib or CIT) plus treatment-free days until treatment switch, end of continuous enrollment, or study end date (03/31/2020), whichever came first.

Study Population

- Inclusion/exclusion criteria are shown in **Figure 2**.
- Patients were classified as receiving either ibrutinib or CIT as 1L CLL treatment, and these groups compared across outcomes reported here.

Study Outcomes Definitions

- All-cause and CLL/SLL-related HRU and costs (payer and patient paid) were evaluated during the follow-up period. Any restart of treatment that occurred after 90 days of discontinuation was considered a second line of therapy and was not considered to be reported in these outcomes.
- CLL/SLL-related HRU and costs were defined as any medical claims associated with CLL/SLL diagnosis regardless of diagnosis position and any pharmacy claims for CLL/SLL medications, including ibrutinib and CIT.

Analyses

- Inverse-probability-of-treatment-weighting (IPTW) was used to account for differences in baseline demographics and clinical characteristics. A threshold of <10% for standardized differences was the criterion to consider covariates balanced.[5]
- IPTW-adjusted all-cause and CLL/SLL-related HRU and total costs (pharmacy+ medical) were compared between ibrutinib and CIT cohorts using a generalized linear model with Poisson and gamma distribution, respectively, and log link, offset by the log of follow-up time.
- Mean monthly cost differences (MMCDs) between the two cohorts were reported on a per-patient-per-month (PPPM) basis. Costs were adjusted to 2019 dollars.

RESULTS

Figure 2: Selection of the Study Population

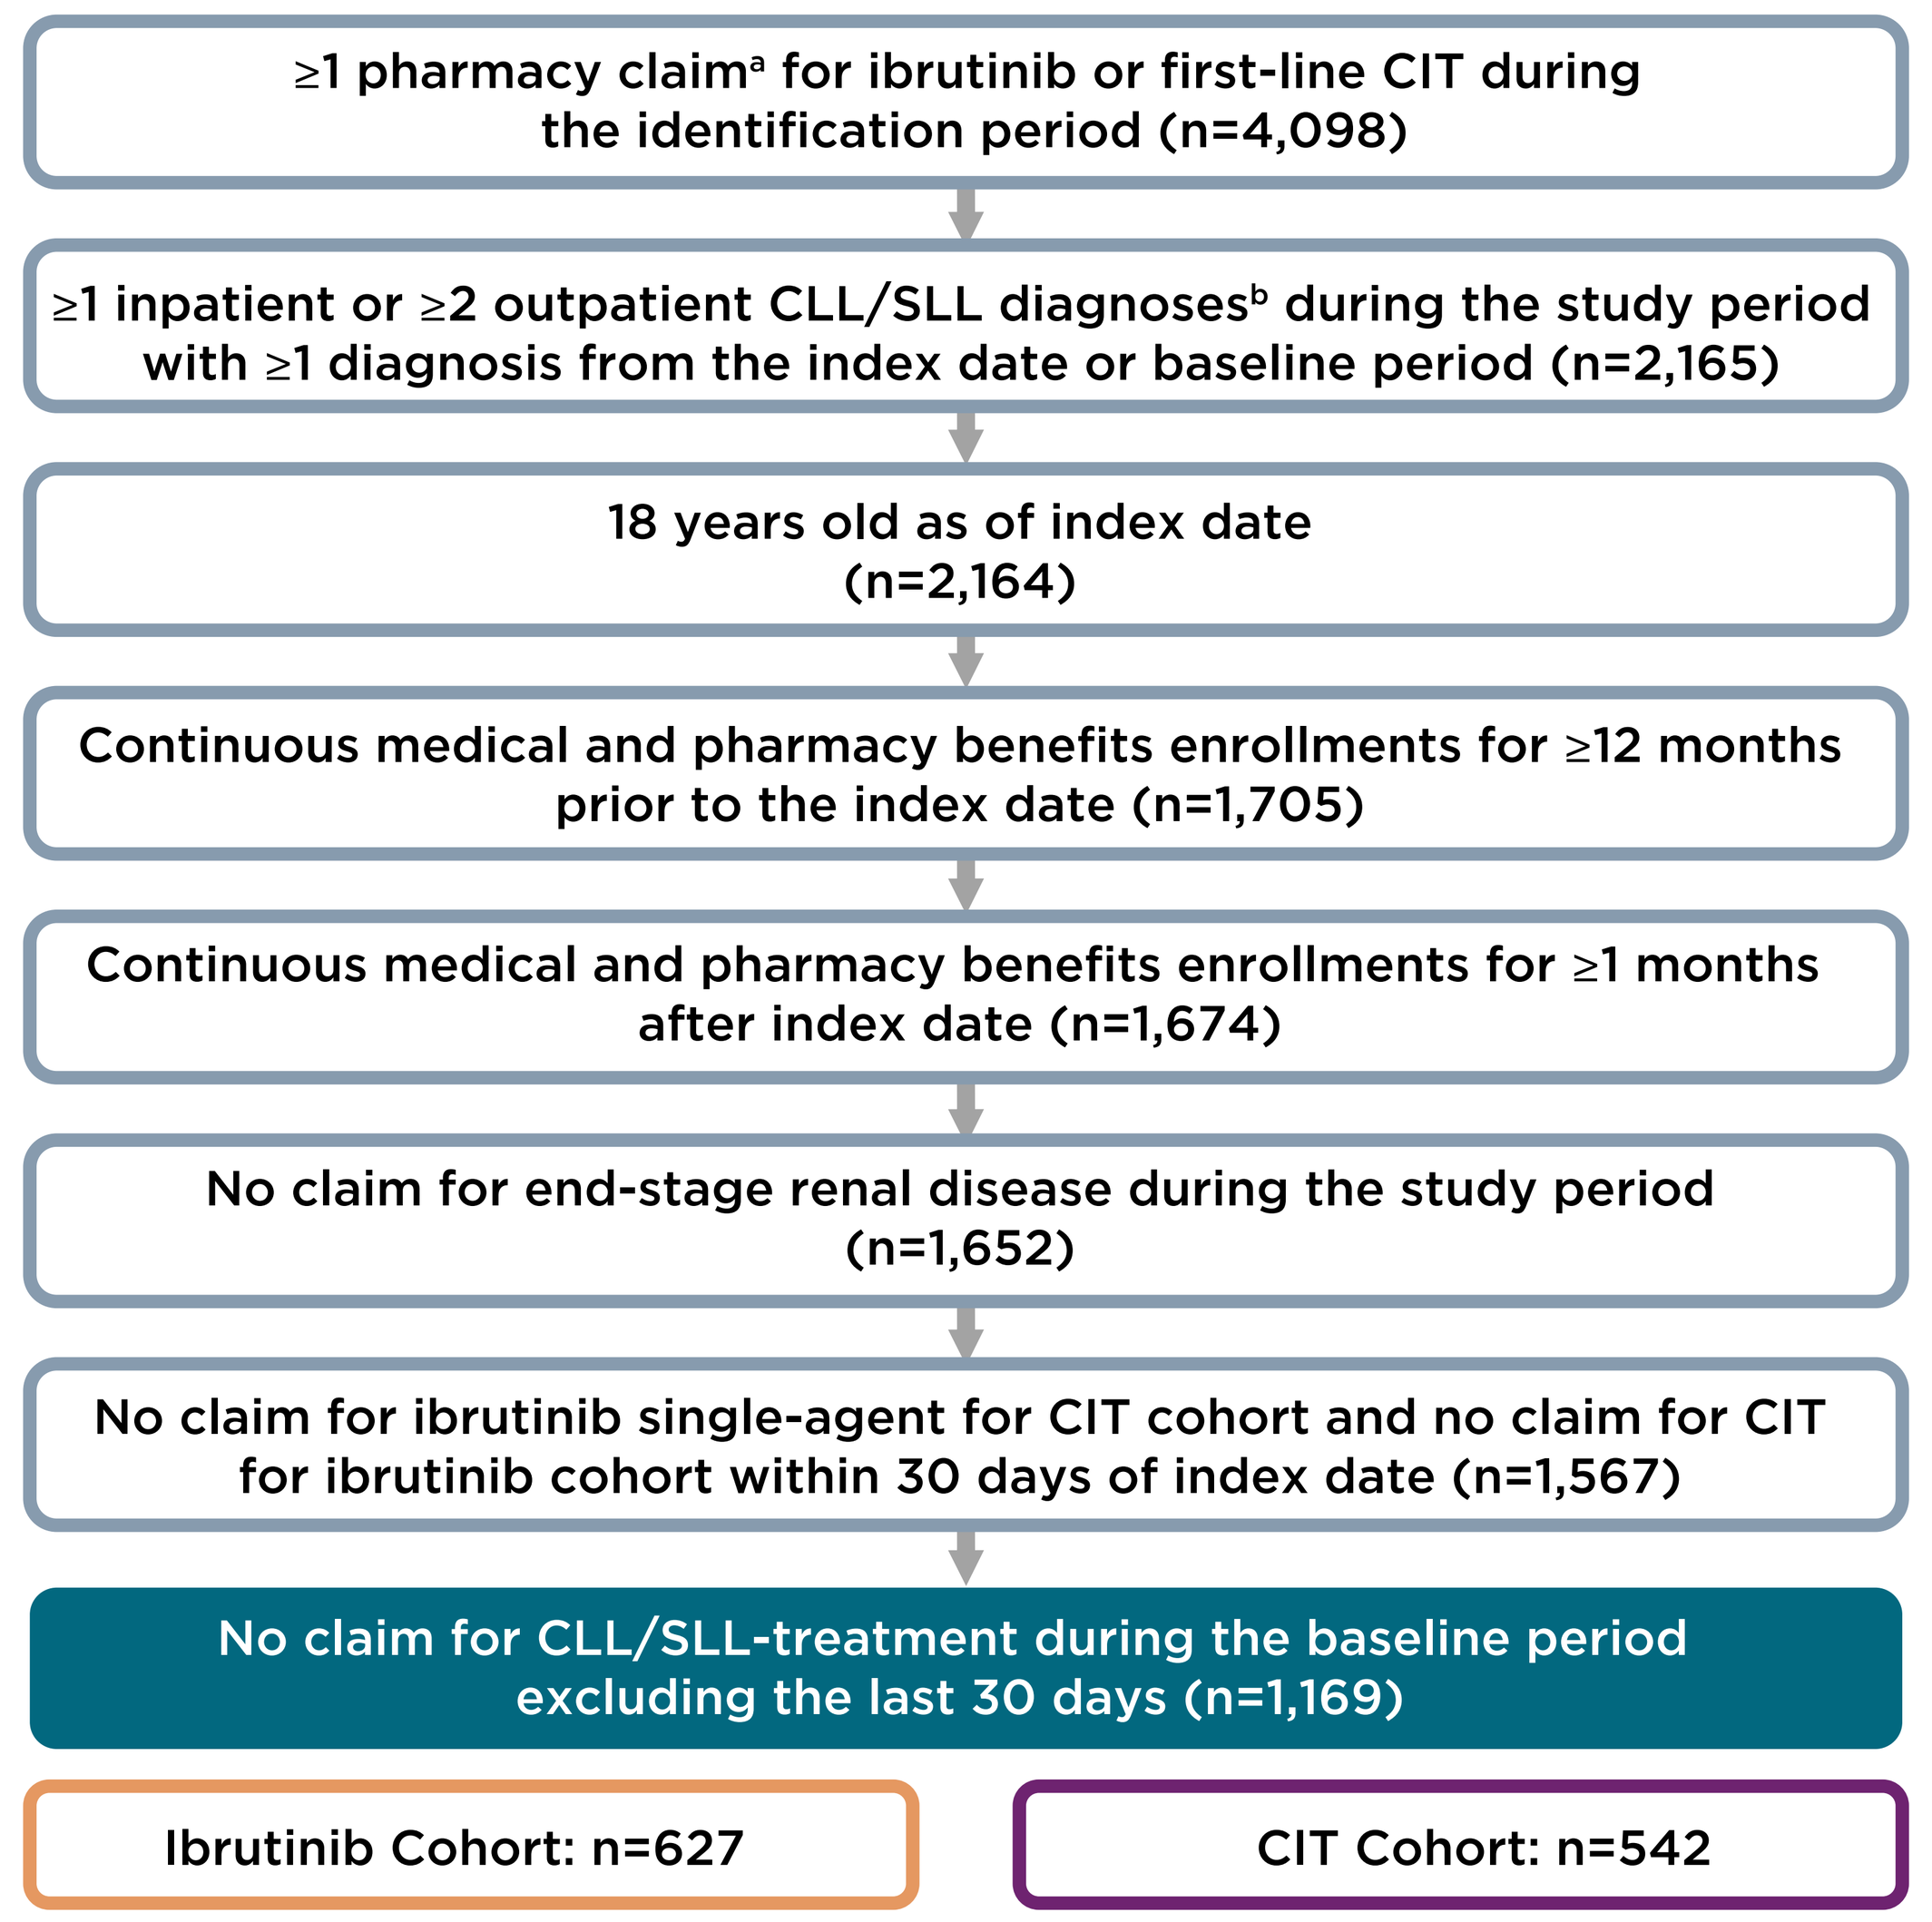
≥1 pharmacy claim[a] for ibrutinib or first-line CIT during the identification period (n=4,098)
≥1 inpatient or ≥2 outpatient CLL/SLL diagnoses[b] during the study period with ≥1 diagnosis from the index date or baseline period (n=2,165)
18 years old as of index date (n=2,164)
Continuous medical and pharmacy benefits enrollments for ≥12 months prior to the index date (n=1,705)
Continuous medical and pharmacy benefits enrollments for ≥1 months after index date (n=1,674)
No claim for end-stage renal disease during the study period (n=1,652)
No claim for ibrutinib single-agent for CIT cohort and no claim for CIT for ibrutinib cohort within 30 days of index date (n=1,567)
No claim for CLL/SLL-treatment during the baseline period excluding the last 30 days (n=1,169)
Ibrutinib Cohort: n=627
CIT Cohort: n=542

[a]Earliest claim for ibrutinib or CIT (bendamustine + obinutuzumab, bendamustine + ofatumumab, bendamustine + rituximab, chlorambucil + obinutuzumab, fludarabine + cyclophosphamide + rituximab, fludarabine + rituximab, or pentostatin + cyclophosphamide + rituximab) during the identification period was defined as the index date. For a treatment to be considered combinational, both treatments should have been given within 30 days of each other.
[b]CLL/SLL was identified using ICD-9 code 204.1x and ICD-10 code C91.1x/C83.0x.

RESULTS (CONT.)

Table 1. Patient Demographics and Clinical Characteristics Before and After IPTW Among 1L Ibrutinib and CIT Cohorts

		Before IPTW			After IPTW		
Demographic and clinical characteristics	**Total**	**Ibrutinib**	**CIT**	**Std. Difference (%)**	**Ibrutinib**	**CIT**	**Std. Difference (%)**
Number of patients, n	1,169	627	542		1,171	1,159	
Mean age (SD), years	67.2 (11.3)	69.3 (11.3)	64.8 (10.8)	40.2	67.2 (15.6)	67.0 (15.6)	1.7
Gender, n (%)							
Female	420 (35.9)	239 (38.1)	181 (33.4)	9.9	414 (35.3)	417 (35.9)	-1.3
Male	749 (64.1)	388 (61.9)	361 (66.6)		757 (64.7)	743 (64.1)	
Residence Region, n (%)[a]							
Midwest	378 (32.3)	209 (33.3)	169 (31.2)	18.5	380 (32.4)	372 (32.1)	0
Northeast	195 (16.7)	98 (15.6)	97 (17.9)		199 (17)	199 (17.1)	
South	316 (27.0)	160 (25.5)	156 (28.8)		306 (26.1)	300 (25.9)	
West	270 (23.1)	154 (24.6)	116 (21.4)		276 (23.6)	277 (23.9)	
Missing	* (*)	* (*)	* (*)		11 (0.9)	11 (1.0)	
Insurance Type,[b] n (%)[a]							
Commercial	655 (56.0)	314 (50.1)	341 (62.9)	26.7	653 (55.8)	655 (56.5)	2.6
Medicare Advantage	274 (23.4)	172 (27.4)	102 (18.8)		272 (23.2)	268 (23.1)	
Medicare Supplemental/Part D	240 (20.5)	141 (22.5)	99 (18.3)		246 (21.0)	236 (20.4)	
Plan type,[c] n (%)[a]							
CDHP	122 (10.4)	56 (8.9)	66 (12.2)	10.9	122 (10.4)	120 (10.4)	0
HMO	336 (28.7)	190 (30.3)	146 (26.9)		331 (28.3)	327 (28.2)	
PPO	711 (60.8)	381 (60.8)	330 (60.9)		718 (61.3)	712 (61.4)	
Year of index date, n (%)[a]							
2016	314 (26.9)	145 (23.1)	169 (31.2)	39.6	310 (26.5)	310 (26.7)	7.3
2017	305 (26.1)	143 (22.8)	162 (29.9)		311 (26.5)	307 (26.5)	
2018	263 (22.5)	141 (22.5)	122 (22.5)		264 (22.5)	257 (22.1)	
2019	247 (21.1)	169 (27.0)	78 (14.4)		248 (21.1)	250 (21.5)	
2020	40 (3.4)	29 (4.6)	11 (2.0)		39 (3.4)	36 (3.1)	
Index treatment prescriber/medication administrator specialty,[d] n (%)							
Oncologist/hematologist	813 (69.5)	558 (89.0)	255 (47.0)	100.7	1,042 (89.0)	581 (50.2)	93.2
Other[e]	356 (30.5)	69 (11.0)	287 (53.0)		129 (11.0)	578 (49.8)	
QCI, n (%)							
0–1	0	0	0	-5.3	0	0	-8.7
0-2	573 (49.0)	307 (49.0)	266 (49.1)		591 (50.5)	570 (49.2)	
≥3	596 (51.0)	320 (51.0)	276 (50.9)		580 (49.5)	589 (50.8)	
CLL-related symptoms, n (%)							
Enlarged lymph nodes[a]	656 (56.6)	326 (52.0)	330 (62.1)	-17.6	660 (56.3)	659 (56.8)	-1.0
Infections[c]	539 (46.5)	297 (47.4)	242 (45.6)	4.0	555 (47.4)	508 (43.8)	7.2
Anemia	476 (41.1)	260 (41.5)	216 (40.7)	2.2	483 (41.2)	473 (40.8)	1.0
Fever/flu-like symptoms	567 (49.0)	314 (50.1)	253 (47.6)	4.6	597 (51.0)	539 (46.5)	9.0
Fatigue/weakness	382 (33.0)	206 (32.9)	176 (33.1)	1.2	387 (33.1)	366 (31.6)	3.1
Splenomegaly[a]	312 (26.9)	156 (24.9)	156 (29.4)	-10.4	303 (25.9)	304 (26.2)	-0.9
Thrombocytopenia	279 (24.1)	150 (23.9)	129 (24.3)	0.7	284 (24.3)	274 (23.6)	1.5
Abdominal pain	251 (21.7)	133 (21.2)	118 (22.2)	-2.3	236 (20.2)	245 (21.1)	-2.4
Other CLL symptoms[f]	341 (29.4)	177 (28.2)	164 (30.9)	-5.3	319 (27.3)	362 (31.2)	-8.7
CLL-related conditions, n (%)							
Hypertension	700 (60.4)	375 (59.8)	325 (61.2)	-1.1	671 (57.3)	715 (61.6)	-8.8
Lymphoma[a]	554 (47.8)	240 (38.3)	314 (59.1)	-43.3	568 (48.5)	572 (49.3)	-1.7
Baseline All-Cause Medical Total Costs, $ PPPM[a]							
Mean (SD)	2,029 (3,682)	1,859 (3,893)	2,226 (3,415)	-10.0	1,954 (5,455)	2,006 (4,728)	-1.0
Demographic and clinical characteristics[g]							
Any malignancy, including leukemia and lymphoma	1,158 (100.0)	627 (100.0)	531 (100.0)	0	1,171 (100.0)	1,159 (100.0)	0
Chronic pulmonary disease	269 (23.2)	137 (21.9)	132 (24.9)	-7.2	272 (23.2)	276 (23.8)	-1.3
Diabetes without chronic complication[a]	267 (23.1)	145 (23.1)	122 (23.0)	10.2	260 (22.2)	292 (25.2)	-0.3
Peripheral vascular disease	156 (13.5)	87 (13.9)	69 (13.0)	2.3	142 (12.1)	157 (13.5)	-4.2
Renal disease[a]	138 (11.9)	88 (14.0)	50 (9.4)	13.8	140 (12.0)	139 (12.0)	0
Mild liver disease	120 (10.4)	64 (10.2)	56 (10.5)	-2.2	126 (10.8)	111 (9.6)	4.0
Cerebrovascular disease	111 (9.6)	66 (10.5)	45 (8.5)	5.7	102 (8.7)	111 (9.5)	-2.8
Congestive heart failure	115 (9.9)	67 (10.7)	48 (9.0)	6.8	116 (9.9)	109 (9.4)	1.8
Diabetes with chronic complication	114 (9.8)	71 (11.3)	43 (8.1)	-0.7	117 (10.0)	117 (10.1)	-7.1
Metastatic solid tumor[a]	73 (6.3)	31 (4.9)	42 (7.9)	-10.8	70 (6.0)	70 (6.1)	-0.5
Myocardial infarction	54 (4.7)	29 (4.6)	25 (4.7)	-0.8	49 (4.2	65 (5.6)	-6.6

In instances where the reported data variables represent small groups (e.g., ≤10), the results are reported as * (*).
[a]Included in IPTW weighting model.
[b]Some patients had more than one plan type, which were recoded into mutually exclusive categories using the hierarchy: Commercial>Medicare Advantage>Medicare Supplement/Part D.
[c]Some patients had more than one type of plan type, which were recoded into mutually exclusive groups using the hierarchy: HMO>PPO>CDHP.
[d]Based on the physician who prescribed the index ibrutinib or CIT medication.
[e]Others include gastroenterology, non-physician clinician, PCP, OB/gynecologist, psychiatrists, neurologists, and other specialty.
[f]Includes atrial fibrillation, weight loss, pancytopenia, hepatomegaly, night sweats, and neutropenia; due to <20% of sample having each condition.
[g]Only conditions occurring in ≥5.0% of patients are shown. The standardized differences were all <10% for these conditions.
QCI, Quan-Charlson Comorbidity Index.

- With the exception of prescriber/medication administrator specialty, baseline demographics and clinical characteristics were well-balanced between cohorts after IPTW adjustments (**Table 1**).

Figure 3. Ibrutinib Demonstrated Lower All-Cause and CLL/SLL-Specific HRU Compared to CIT

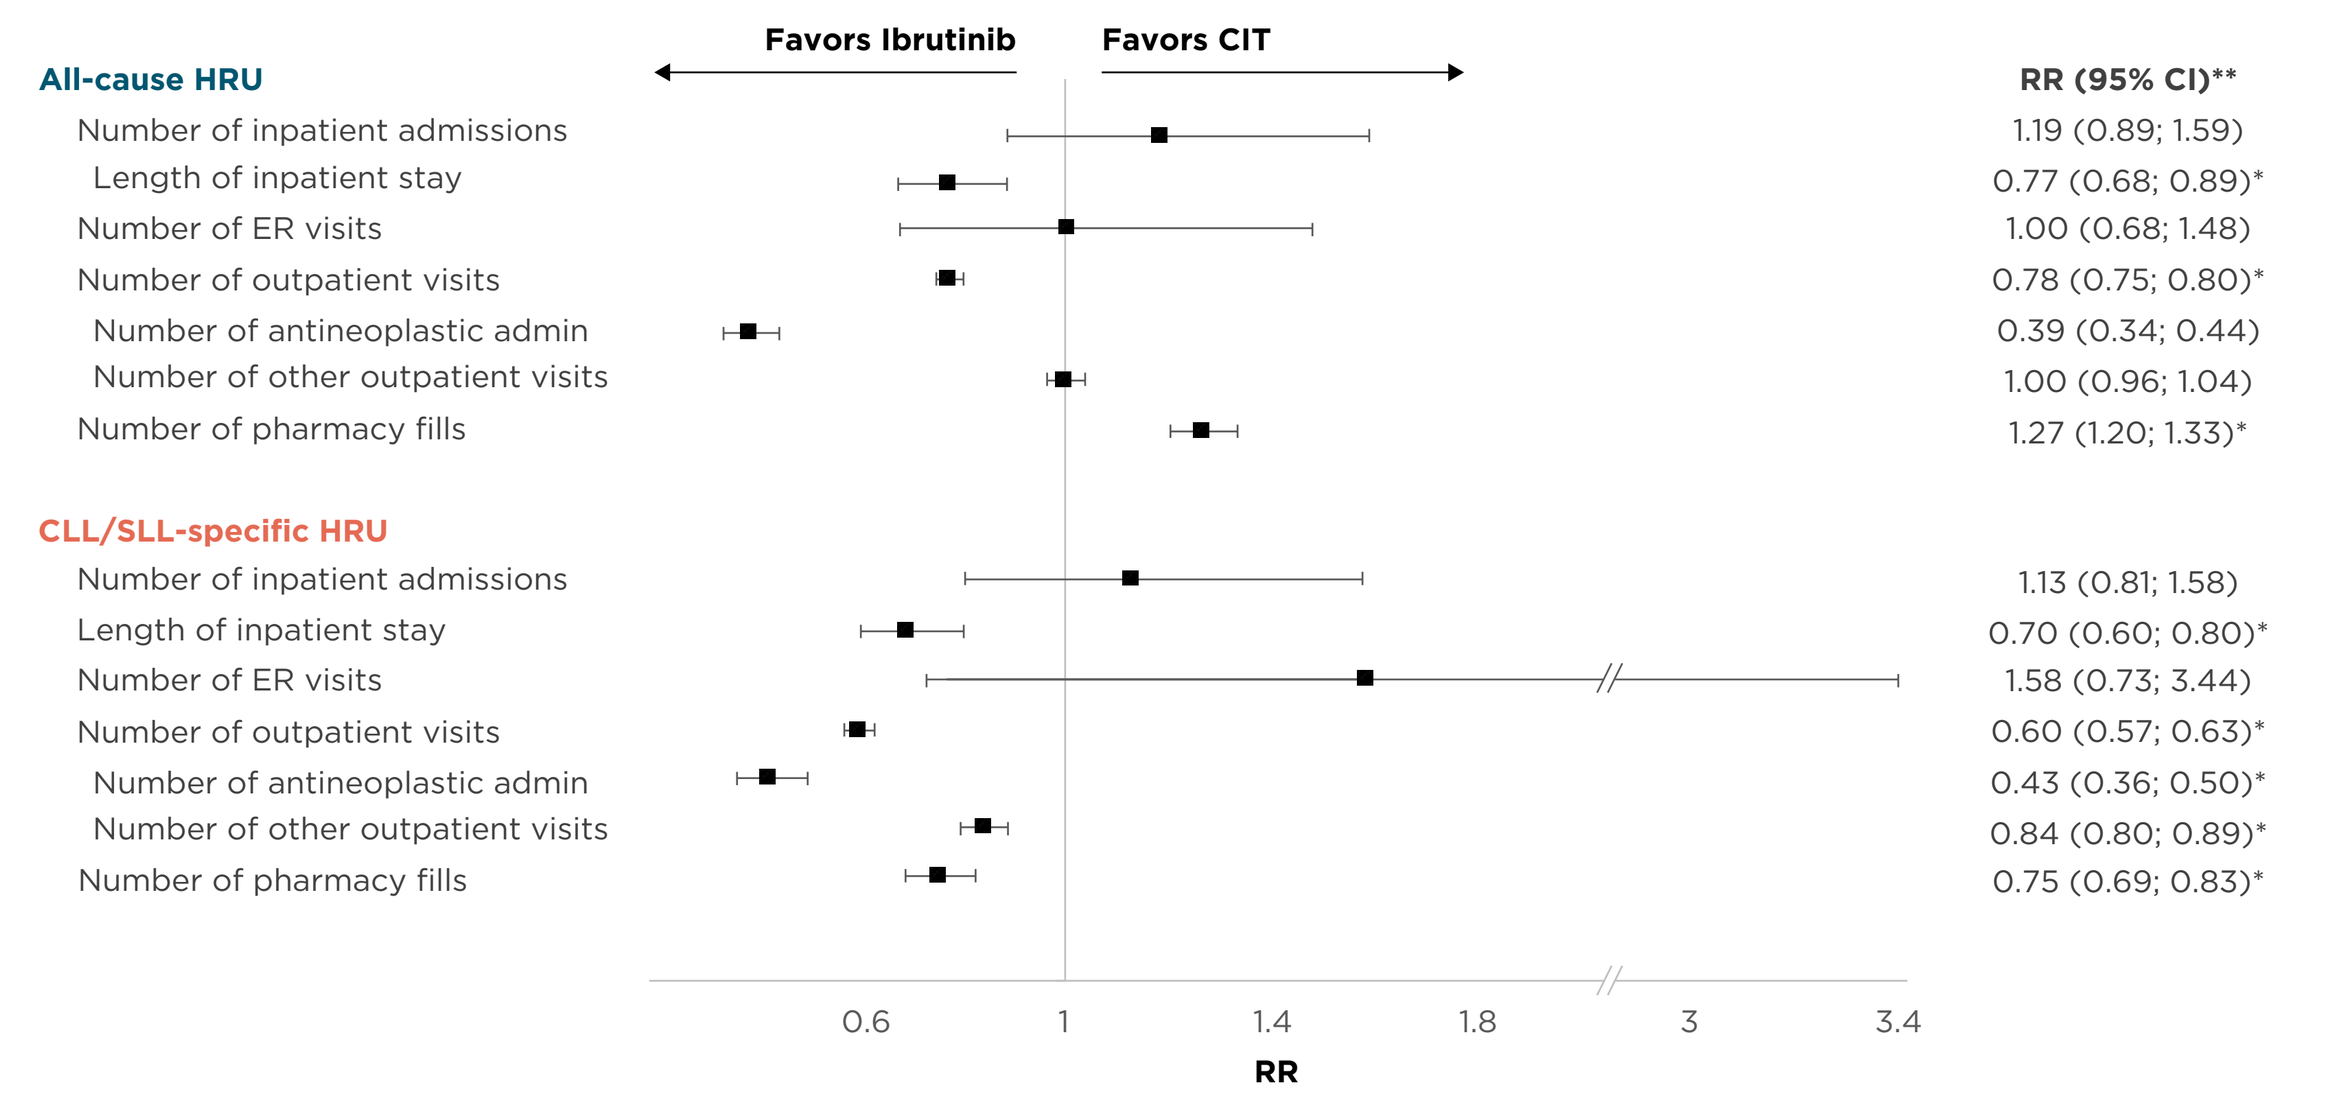
Favors Ibrutinib
Favors CIT
RR (95% CI)**
All-cause HRU
Number of inpatient admissions 1.19 (0.89; 1.59)
Length of inpatient stay 0.77 (0.68; 0.89)*
Number of ER visits 1.00 (0.68; 1.48)
Number of outpatient visits 0.78 (0.75; 0.80)*
Number of antineoplastic admin 0.39 (0.34; 0.44)
Number of other outpatient visits 1.00 (0.96; 1.04)
Number of pharmacy fills 1.27 (1.20; 1.33)*
CLL/SLL-specific HRU
Number of inpatient admissions 1.13 (0.81; 1.58)
Length of inpatient stay 0.70 (0.60; 0.80)*
Number of ER visits 1.58 (0.73; 3.44)
Number of outpatient visits 0.60 (0.57; 0.63)*
Number of antineoplastic admin 0.43 (0.36; 0.50)*
Number of other outpatient visits 0.84 (0.80; 0.89)*
Number of pharmacy fills 0.75 (0.69; 0.83)*
0.6
1
1.4
1.8
3
3.4
RR

*P<0.05.
**RRs were calculated using a generalized linear model with normal distribution, weighted for IPTW. RRs used CIT as the reference cohort.
†Other outpatient services include outpatient office visits, procedures, laboratory tests, imaging, other tests, physical therapy, occupational therapy, speech therapy, and other physician services, including behavioral therapy.
ER, emergency room; RR, rate ratio.

- Ibrutinib-treated patients had significantly more pharmacy visits relative to CIT (RR, 1.27; *P*<0.001).However, ibrutinib-treated patients had significantly fewer days of all-cause outpatient visits compared to CIT-treated patients (RR, 0.78; *P*<0.001; **Figure 3**).
- Ibrutinib patients had significantly shorter inpatient lengths of stay (RR, 0.77; **Figure 3**).

Figure 4. Ibrutinib Demonstrated Lower All-Cause and CLL/SLL-Specific Costs PPPM Compared to CIT

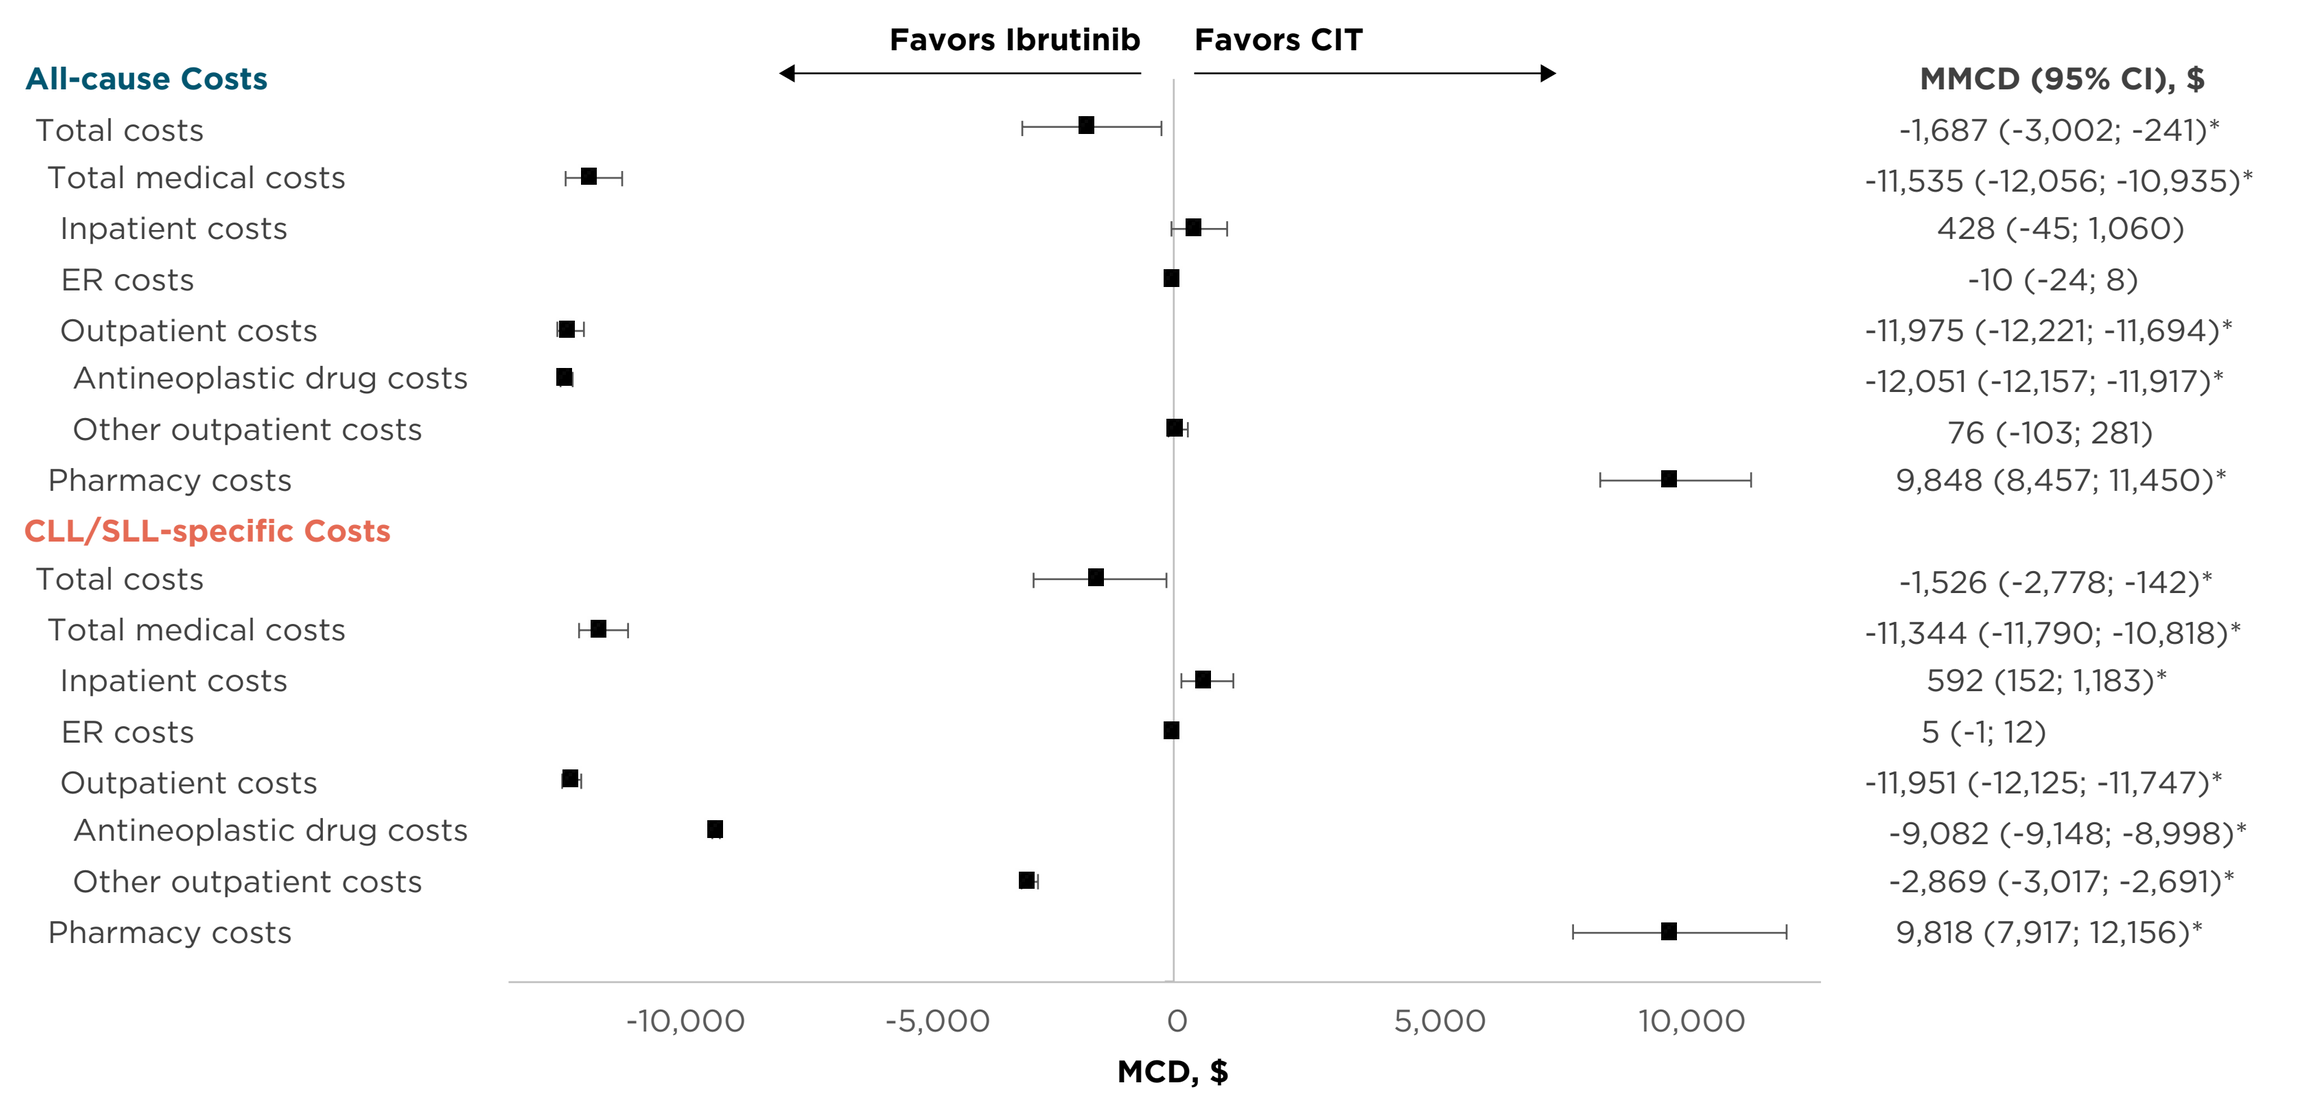
Favors Ibrutinib
Favors CIT
MMCD (95% CI), $
All-cause Costs
Total costs -1,687 (-3,002; -241)*
Total medical costs -11,535 (-12,056; -10,935)*
Inpatient costs 428 (-45; 1,060)
ER costs -10 (-24; 8)
Outpatient costs -11,975 (-12,221; -11,694)*
Antineoplastic drug costs -12,051 (-12,157; -11,917)*
Other outpatient costs 76 (-103; 281)
Pharmacy costs 9,848 (8,457; 11,450)*
CLL/SLL-specific Costs
Total costs -1,526 (-2,778; -142)*
Total medical costs -11,344 (-11,790; -10,818)*
Inpatient costs 592 (152; 1,183)*
ER costs 5 (-1; 12)
Outpatient costs -11,951 (-12,125; -11,747)*
Antineoplastic drug costs -9,082 (-9,148; -8,998)*
Other outpatient costs -2,869 (-3,017; -2,691)*
Pharmacy costs 9,818 (7,917; 12,156)*
-10,000
-5,000
0
5,000
10,000
MCD, $

*P<0.05.
**MMCDs were calculated using a generalized linear model with gamma distribution and log link, weighted for IPTW. Outcomes modeled as PPPM costs to adjust for variation in the follow-up.
†Other outpatient services include outpatient office visits, procedures, laboratory tests, imaging, other tests, physical therapy, occupational therapy, speech therapy, and other physician services, including behavioral therapy.

- Ibrutinib-treated patients incurred lower all-cause medical cost (MMCD, $11,535; *P*<0.001) and higher all-cause pharmacy cost (MMCD, $9,848; *P*<0.001), resulting in net savings of $1,687 (*P*<0.001) PPPM associated with ibrutinib compared to CIT (**Figure 4**).
 - The higher medical cost among CIT-treated patients was due to significantly higher antineoplastic drug-related cost in the outpatient setting (MMCD, -$12,051; *P*<0.001).
- CLL-specific outcomes (HRU and cost) were directionally consistent with all-cause outcomes (**Figures 3 and 4**).

DISCUSSION/CONCLUSIONS

- Total all-cause and CLL-specific direct costs of care were lower with ibrutinib vs CIT in 1L treatment of CLL (all-cause total costs MMCD, -$1,687; CLL/SLL-specific total costs MMCD, -$1,526), resulting from higher pharmacy costs of ibrutinib being fully offset by lower medical costs associated with ibrutinib; this was driven mostly by fewer outpatient visits and associated costs.
- Outpatient utilization and costs among patients treated with ibrutinib were observed to be significantly lower than those among CIT-treated patients (number of outpatient visits RR, 0.60; outpatient costs MMCD, -$11,975).
- Because CIT is primarily covered as a medical benefit, greater outpatient service utilization and costs among CIT-treated patients were driven by significantly higher chemotherapy administration and other outpatient costs associated with chemotherapy (antineoplastic administration RR and costs, 0.39 and -$12,051, respectively; other outpatient visits RR and costs, 1.0 and $76, respectively).
- Alternatively, since ibrutinib is covered as a pharmacy benefit, pharmacy costs among ibrutinib patients were observed to be significantly higher than those among CIT-treated patients (pharmacy costs MMCD, $9,848).
- Ibrutinib patients had significantly shorter all-cause inpatient lengths of stay (RR, 0.77).

LIMITATIONS

- Claims data may contain omissions and inaccuracies, but this is expected to equally affect all cohorts, and thus, should not impact the overarching conclusions of this study.
- The 12-month baseline period was used to identify 1L ibrutinib and CIT users among the CLL population. If a patient took a treatment holiday of longer than 12 months, this may have falsely classified the patient as a newly treated patient, resulting in misclassification bias.
- Mutational status is unavailable in claims data. It is well-established that ibrutinib compared with CIT works well in the treatment of del(17p)/*TP53* mutation among patients with CLL/SLL.[4,8] Such aberrations are associated with an unfavorable prognosis[9]; hence, conclusions of the current study are likely conservative estimates of the potential cost savings associated with ibrutinib treatment relative to CIT.

REFERENCES
1. National Cancer Institute SEER Program. 2017. https://seer.cancer.gov/statfacts/html/clyl.html.
2. Byrd JC, et al. *N Engl J Med.* 2014;371(3):213-23.
3. Burger JA, et al. *N Engl J Med.* 2015;373(25):2425-37.
4. Moreno C, et al. *Lancet Oncol.* 2019;20(1):43-56.
5. Woyach JA, et al. *N Engl J Med.* 2018;379(26):2517-28.
6. Shanafelt TD, et al. *N Engl J Med.* 2019;381(5):432-43.
7. Fraser G, et al. *Leukemia.* 2019;33(4):969-80.
8. Munir T, et al. *Am J Hematol.* 2019;94(12):1353-63.
9. Stilgenbauer S, et al. *Blood.* 2014;123(21):3247-54.

DISCLOSURES
RRV and SC: employment with Pharmacyclics LLC, an AbbVie Company; stock ownership in AbbVie; **RI:** employment with Pharmacyclics LLC, an AbbVie Company; stock ownership in AbbVie; patents, royalties, or other intellectual property with Express Scripts; **MS:** employment with HealthCore Inc.; stock ownership in Anthem; research funding from Pharmacyclics LLC, an AbbVie Company; **JLS:** employment with HealthCore, Inc.; stock ownership in Anthem; research funding from Pharmacyclics LLC, an AbbVie Company.

ACKNOWLEDGMENTS
This study was sponsored by Pharmacyclics LLC, an AbbVie Company. Editorial support was provided by Meghan Sullivan, PhD, CMPP, and funded by Pharmacyclics LLC, an AbbVie Company.

Presented at the Academy of Managed Care Pharmacy (AMCP) Annual Meeting, April 13-16, 2021, San Francisco, CA